P-2036

OAKTREE

Obeldesivir for Treatment of COVID-19 in Adults and Adolescents Without Risk Factors for Progression to Severe Disease: The OAKTREE Study

Onyema Ogbuagu[1], Jason D Goldman[2,3], Robert L Gottlieb[4,5], Upinder Singh[6], Masaharu Shinkai[7], Gerard Acloque[8], Dahlene N Fusco[9], Erika Gonzalez[10], Princy Kumar[11], Anne Luetkemeyer[12], Amos Lichtman[13], Afsaneh Mozaffarian[13], Yiannis Koullias[13], Robert H Hyland[13], Joe Llewellyn[13], Anu Osinusi[13], Frank Duff[13], Rita Humeniuk[13], Luzelena Caro[13], Santosh Davies[13], Charlotte Hedskog[13], Shuguang Chen[13], Kim Etchevers[13], Priyanka Nadig[13], Anita Kohli[14,15]

[1]Yale School of Medicine, New Haven, CT, USA; [2]Providence Swedish Medical Center, Seattle, WA, USA; [3]University of Washington, Seattle, WA, USA; [4]Baylor University Medical Center, Dallas, TX, USA; [5]Baylor Scott & White Research Institute, Dallas, TX, USA; [6]Stanford Medicine, Stanford, CA, USA; [7]Tokyo Shinagawa Hospital, Tokyo, Japan; [8]Universal Medical and Research Center, LLC, Coral Gables, FL, USA; [9]Tulane University School of Medicine, New Orleans, LA, USA; [10]STAAMP Research, San Antonio, TX, USA; [11]Georgetown University Medical Center, Washington DC, USA; [12]University of California San Francisco, San Francisco, CA, USA; [13]Gilead Sciences, Inc., Foster City, CA, USA; [14]Arizona Clinical Trials, Chandler, AZ, USA; [15]Arizona Liver Health, Tucson, AZ, USA

Onyema Ogbuagu
onyema.ogbuagu@yale.edu

Copies of this poster obtained through QR (Quick Response) are for personal use only and may not be reproduced without written permission of the authors

Conclusions

- In nonhospitalized adults and adolescents without risk factors for progression to severe disease, obeldesivir 350 mg administered twice daily for 5 days was generally safe and well tolerated but did not significantly reduce time to COVID-19 symptom alleviation or resolution compared to placebo
- Obeldesivir treatment reduced SARS-CoV-2 viral RNA copy number at Days 3 and 5 and resulted in a greater proportion of participants who tested negative for SARS-CoV-2 at Day 5 compared to placebo
- COVID-19 symptom duration was markedly shorter than in earlier trials, consistent with milder disease in a population with high rates of hybrid immunity during the Omicron era
- Obeldesivir remains a promising treatment for COVID-19 and other coronavirus-mediated diseases given its demonstrated antiviral activity and favorable safety profile

Plain Language Summary

- Obeldesivir is an oral medication that stops the replication of SARS-CoV-2, the virus that causes COVID-19
- This study aimed to find out if obeldesivir was safe and effective for treating COVID-19 in people at low risk of becoming severely ill from the disease
- There were no major differences in the time to COVID-19 symptom improvement when people took obeldesivir compared to placebo
- This may have been because most people in the study already had some immunity to the virus and got better quickly, even without treatment
- Obeldesivir was generally safe and well tolerated, and almost all people completed their assigned treatment
- Obeldesivir also lowered the amount of SARS-CoV-2 virus a person had compared to placebo
- Overall, obeldesivir remains a promising treatment for COVID-19 and other similar illnesses

Introduction

- COVID-19 remains a public health priority, with a higher annual mortality than influenza[1-3]
- There are currently no antiviral treatment options approved in the United States for mild-to-moderate COVID-19 for nonhospitalized individuals without risk factors for progression to severe disease[4]
- Obeldesivir (ODV) is an oral nucleoside analog prodrug that, through a different pharmacometabolic pathway, delivers the same active metabolite as remdesivir that inhibits SARS-CoV-2 RNA-dependent RNA polymerase[5,6]
- ODV has a low pill burden, no clinically meaningful drug-drug interactions, and in vitro antiviral activity against SARS-CoV-2 variants[5,7-10]

Objective

- To evaluate the efficacy, safety, and tolerability of ODV for the treatment of COVID-19 in nonhospitalized individuals without risk factors for progression to severe disease

Methods

- OAKTREE (ClinicalTrials.gov Identifier: NCT05715528) was a Phase 3, randomized, double-blind, placebo-controlled study in nonhospitalized participants aged ≥12 to <65 years without specified risk factors for progression to severe COVID-19
- Participants were enrolled (February 13-October 31, 2023) within 3 days of the onset of ≥2 targeted COVID-19 symptoms (stuffy or runny nose, sore throat, shortness of breath [difficulty breathing], cough, low energy or tiredness, muscle or body aches, headache, chills or shivering, and feeling hot or feverish), stratified by completion of a primary COVID-19 vaccine series, and randomized 1:1 to receive ODV 350 mg or placebo twice daily for 5 days
- Primary endpoints were time to COVID-19 symptom alleviation by Day 29, incidence of treatment-emergent adverse events (AEs) and laboratory abnormalities, and incidence of serious AEs and AEs leading to study drug discontinuation
 — Symptom alleviation occurred if, for ≥48 consecutive hours, all targeted symptoms scored as moderate or severe at baseline were scored as mild or none and all targeted symptoms scored as mild or none at baseline were scored as none
- Secondary endpoints included time to COVID-19 symptom resolution by Day 29, change from baseline in SARS-CoV-2 nasal swab viral RNA copy number, and COVID-19–related hospitalization or all-cause death by Day 29

Results

Participants

- Overall, 1955 participants (ODV, 979; placebo, 976) were randomized and received ≥1 dose of study drug, of whom 1924 (98%) completed study drug
- Baseline characteristics were generally similar between groups (Table 1)
 — Overall, 70% of participants had completed a primary vaccination series, 99% were positive for SARS-CoV-2 anti-spike antibodies, 86% were positive for anti-nucleocapsid antibodies, and the mean baseline SARS-CoV-2 viral RNA copy number was 5.1 $\log_{10}$ copies/mL

Table 1. Demographic and Baseline Characteristics (Safety Analysis Set[a])

Characteristic	ODV (n = 979)	Placebo (n = 976)	Total (n = 1955)
Age, years, median (range)	42 (12-64)	40 (12-64)	41 (12-64)
Sex at birth, n (%)			
Female	583 (60)	572 (59)	1155 (59)
Race, n (%)			
Asian	21 (2)	21 (2)	42 (2)
Black	106 (11)	101 (10)	207 (11)
White	850 (87)	848 (87)	1698 (87)
Other[b]	2 (<1)	6 (1)	8 (<1)
Ethnicity, n (%)			
Hispanic or Latino	918 (94)	901 (92)	1819 (93)
Completed primary vaccination series, n (%)			
Yes	687 (70)	681 (70)	1368 (70)
COVID-19 vaccination status, n (%)			
Never	271 (28)	266 (27)	537 (27)
Duration of COVID-19 symptoms prior to first dose of study drug, days, median (Q1, Q3)[c]	2 (2, 2)	2 (2, 2)	2 (2, 2)
Participants with COVID-19 symptom data available at baseline, n	962	962	1924
Number of targeted COVID-19 symptoms at baseline, median (Q1, Q3)	8 (6, 9)	7 (6, 9)	8 (6, 9)
Serostatus, n (%)[d]			
Overall positive	970 (>99)	968 (99)	1938 (>99)
Overall negative	1 (<1)	7 (1)	8 (<1)
Missing	8	1	9
SARS-CoV-2 anti-spike antibody, n[e]	884	884	1768
Positive, n (%)	869 (>99)	869 (99)	1738 (99)
Negative, n (%)	3 (<1)	7 (1)	10 (1)
Missing, n	12	8	20
SARS-CoV-2 anti-nucleocapsid antibody, n[e]	884	884	1768
Positive, n (%)	755 (86)	756 (86)	1511 (86)
Negative, n (%)	125 (14)	121 (14)	246 (14)
Missing, n	4	7	11
SARS-CoV-2 viral RNA copy number, n[f]	913	911	1824
$\log_{10}$ copies/mL, mean (SD)	5.1 (1.4)	5.1 (1.5)	5.1 (1.4)

[a]Safety analysis set included participants who were randomized and received ≥1 dose of study drug. Participants in the safety analysis set were summarized according to the actual treatment received.
[b]For the race category, "other" includes American Indian or Alaska Native, Native Hawaiian or Pacific Islander, other, and "not permitted."
[c]Duration of COVID-19 symptoms was defined as the first dosing date minus the COVID-19 symptom onset date (Day 0).
[d]Serostatus was defined as positive when anti-spike antibody or anti-nucleocapsid antibody was positive and was defined as negative when both were negative. Serostatus percentages do not include those with missing values.
[e]Evaluated in the full analysis positive set (participants who were randomized, received ≥1 dose of study drug, and were SARS-CoV-2 positive at baseline as confirmed by RT-PCR at the central laboratory). Percentages do not include those with missing values.
[f]Result of "No SARS-CoV-2 detected" was imputed as 746.5 copies/mL (2.87 $\log_{10}$ copies/mL); "<2228 copies/mL" was imputed as 1114 copies/mL (3.05 $\log_{10}$ copies/mL).
ODV, obeldesivir; RT-PCR, reverse transcriptase–polymerase chain reaction.

Clinical Efficacy

- Of the 1955 participants who were randomized and received ≥1 dose of study drug, 1768 (90%) participants (884 per group) had reverse transcriptase–polymerase chain reaction–confirmed SARS-CoV-2 infection at baseline
- Among those with COVID-19 symptom data, the Kaplan-Meier estimate of median time to symptom alleviation was 5.9 (95% CI, 5.4-6.1) days in the ODV group and 6.0 (95% CI, 5.8-6.3) days in the placebo group (P = 0.068; Figure 1)

Figure 1. Kaplan-Meier Estimate of Time to COVID-19 Symptom Alleviation (Full Analysis Positive Set[a])

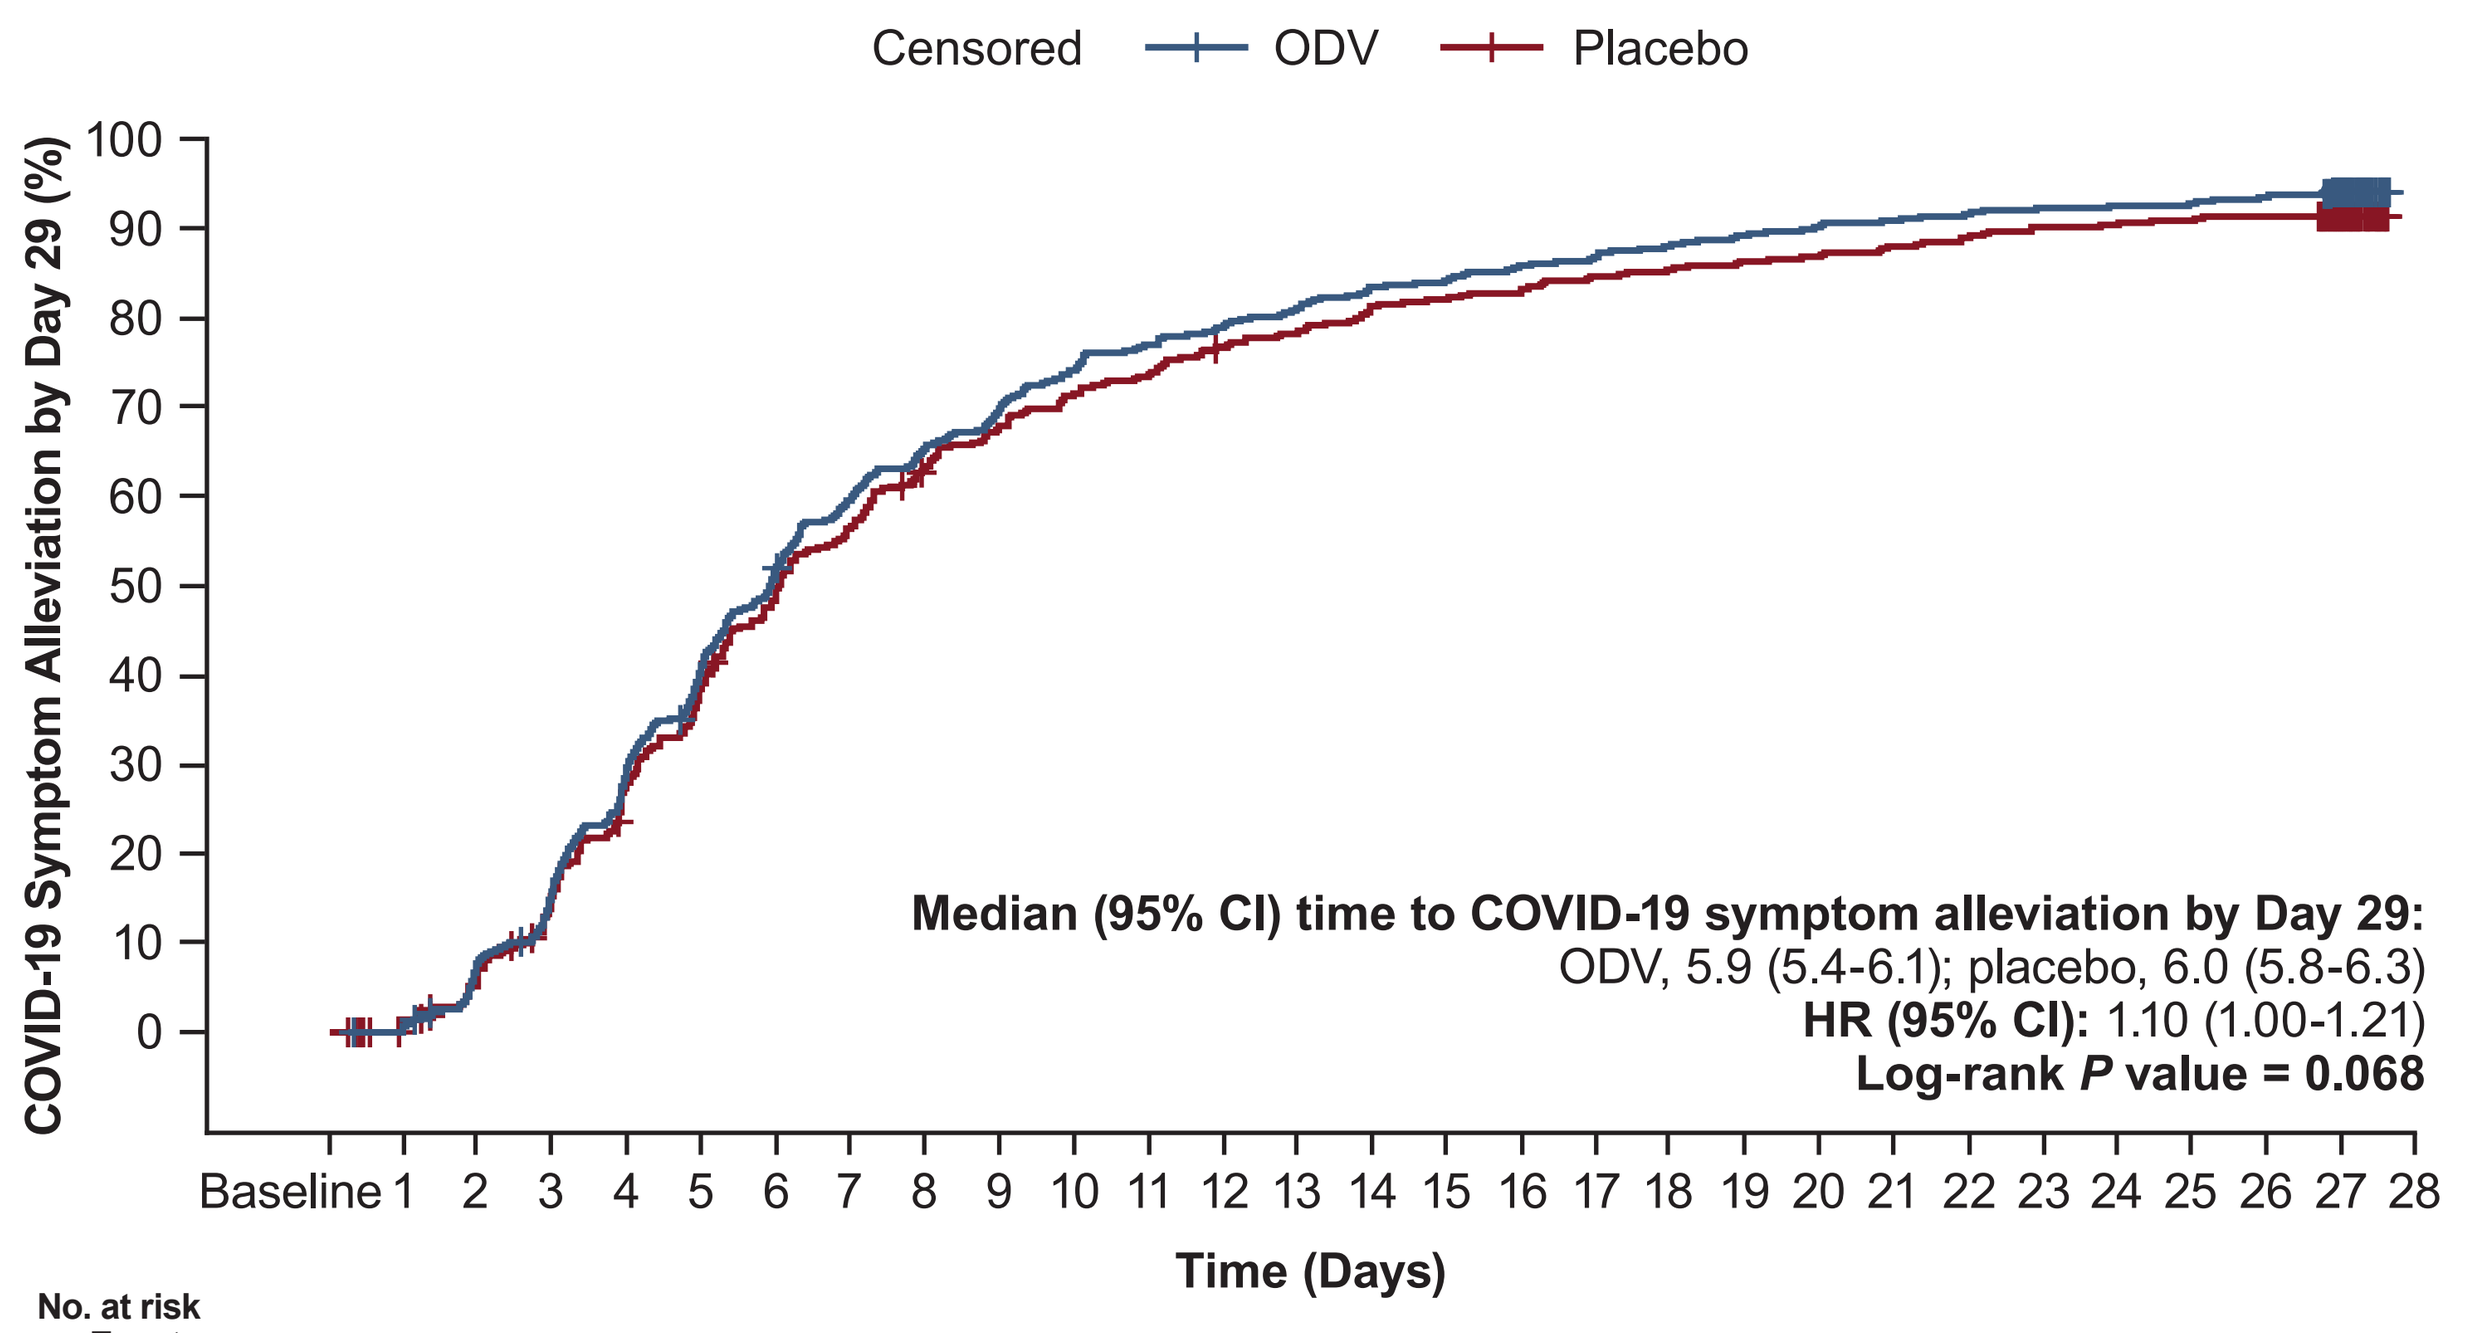

Censored
ODV
Placebo
COVID-19 Symptom Alleviation by Day 29 (%)
100
90
80
70
60
50
40
30
20
10
0
Median (95% CI) time to COVID-19 symptom alleviation by Day 29:
ODV, 5.9 (5.4-6.1); placebo, 6.0 (5.8-6.3)
HR (95% CI): 1.10 (1.00-1.21)
Log-rank P value = 0.068
Baseline 1 2 3 4 5 6 7 8 9 10 11 12 13 14 15 16 17 18 19 20 21 22 23 24 25 26 27 28
Time (Days)

No. at risk / Events	Baseline	1	2	3	4	5	6	7	8	9	10	11	12	13	14	15	16	17	18	19	20	21	22	23	24	25	26	27	28
ODV	879	873	810	739	619	522	420	352	302	263	224	199	183	164	144	137	122	114	105	94	85	79	73	67	64	62	56	32	
	0	4	66	136	256	352	454	521	571	610	649	674	690	709	729	736	751	759	768	779	788	794	800	806	809	811	817	822	822
Placebo	882	872	821	744	629	535	442	379	319	279	247	230	199	187	163	154	145	133	126	117	112	103	94	85	82	75	74	48	
	0	2	52	128	242	336	428	491	549	589	621	638	668	680	704	713	722	734	741	750	755	764	773	782	785	792	793	794	794

Patient-reported outcomes were collected through a COVID-19 symptom questionnaire adapted from US Food and Drug Administration guidance that participants completed daily through Day 29 and on Days 60 and 90. Participants who prematurely discontinued the study prior to Day 29 or whose alleviation status was missing were censored at the last date/time at which the symptom was assessed or Day 28, whichever occurred first. HR and 2-sided 95% CI for HR were estimated using the Cox regression with the randomization stratification factor as a covariate. P value was calculated from stratified log-rank test with the randomization stratification factor as the stratum.
[a]Full analysis positive set included participants who were randomized, received ≥1 dose of study drug, and were SARS-CoV-2 positive at baseline as confirmed by RT-PCR at the central laboratory.
HR, hazard ratio; ODV, obeldesivir; RT-PCR, reverse transcriptase–polymerase chain reaction.

- The Kaplan-Meier estimates of median time to symptom resolution were also similar between treatment groups (ODV, 9.2 [95% CI, 8.9-10.0] days; placebo, 9.3 [95% CI, 8.9-10.1] days; nominal P = 0.556)
- No COVID-19–related hospitalizations or deaths were reported by Day 29 in either group
- No meaningful differences were seen between groups for symptoms at Day 90

Virologic Efficacy

- In the 1478 participants (ODV, 751; placebo, 727) in the virology analysis set, viral RNA copy number decreased rapidly from baseline to Day 5 in both groups (Figure 2)
 — Compared to the placebo group, there were greater reductions in least squares mean viral RNA copy number from baseline to Day 3 (treatment difference [95% CI], –0.31 [–0.46 to –0.16] $\log_{10}$ copies/mL; nominal P <0.0001) and to Day 5 (treatment difference [95% CI], –0.18 [–0.30 to –0.06] $\log_{10}$ copies/mL; nominal P = 0.004) in the ODV group

Figure 2. Least Squares Mean (95% CI) Change From Baseline in SARS-CoV-2 Nasal Swab Viral RNA Copy Number ($\log_{10}$ Copies/mL) Using MMRM (Virology Analysis Set[a])

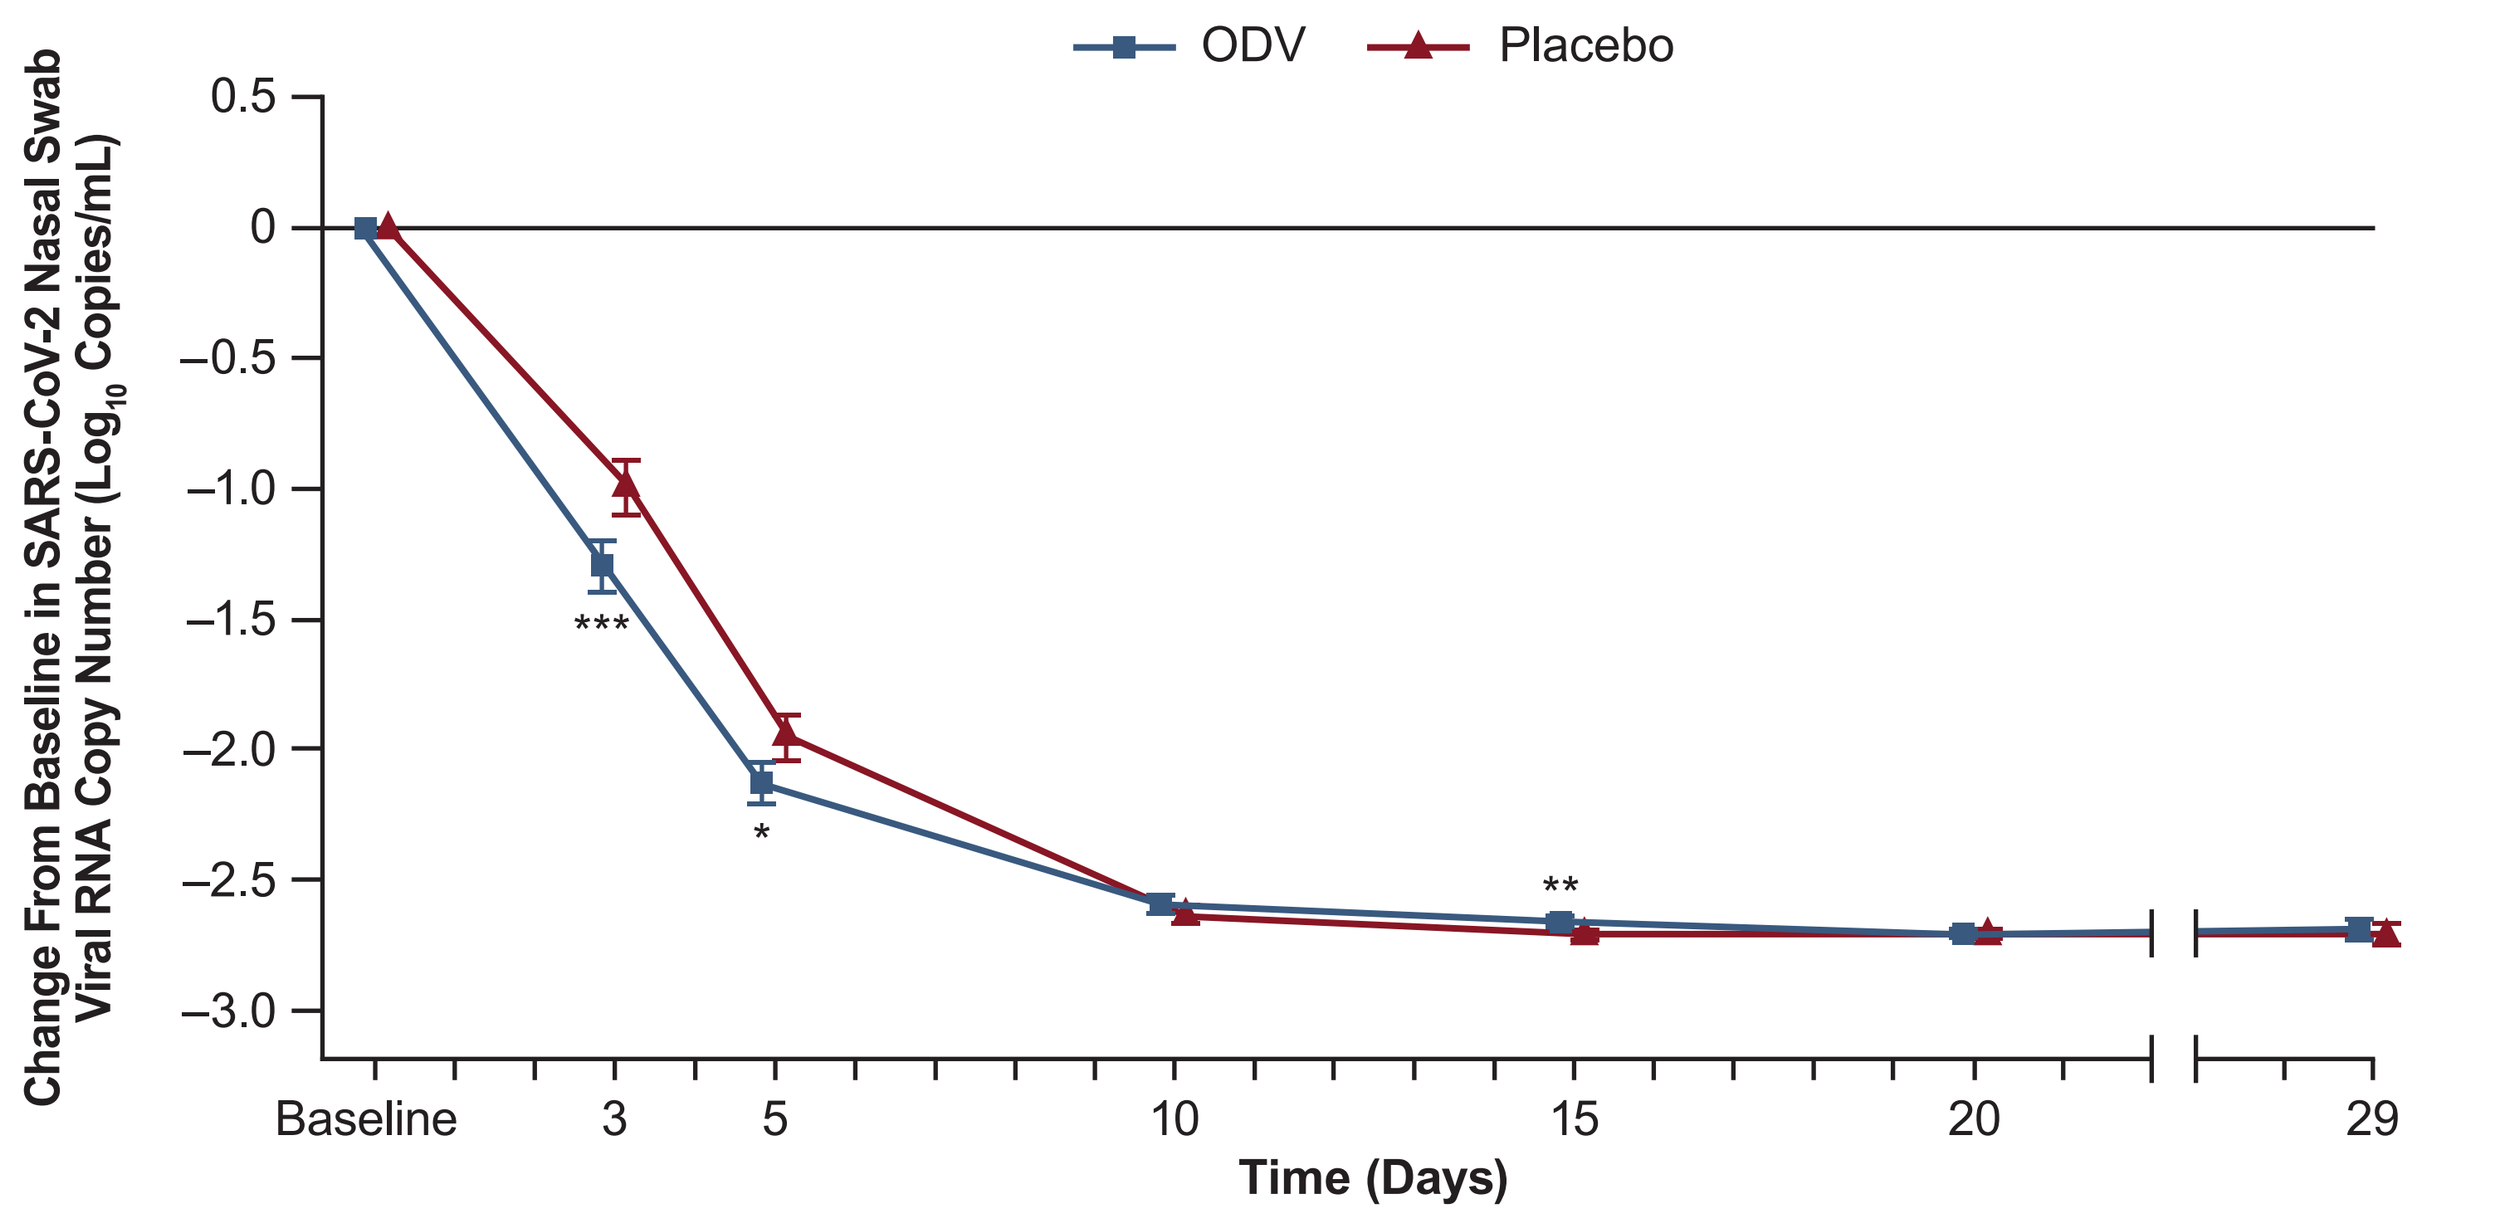

ODV
Placebo
Change From Baseline in SARS-CoV-2 Nasal Swab Viral RNA Copy Number (Log10 Copies/mL)
0.5
0
–0.5
–1.0
–1.5
–2.0
–2.5
–3.0

*
**
Baseline
3
5
10
15
20
29
Time (Days)

	Baseline	3	5	10	15	20	29
ODV (n)	751	656	637	640	644	658	222
Placebo (n)	727	641	622	620	629	644	209

Mean (SE), 95% CI, and P value were from MMRM with baseline viral RNA copy number and the randomization stratum as covariates. * indicates nominal P <0.01 versus placebo. ** indicates nominal P <0.001 versus placebo. *** indicates nominal P <0.0001 versus placebo. The LLOD and LLOQ for SARS-CoV-2 viral RNA were 1493 and 2228 copies/mL, respectively. The result of "No SARS-CoV-2 detected" was imputed as half of the LLOD (746.5 copies/mL; 2.87 $\log_{10}$ copies/mL); the result of "<2228 copies/mL" was imputed as half of the LLOQ (1114 copies/mL; 3.05 $\log_{10}$ copies/mL).
[a]Virology analysis set included participants who were randomized, received ≥1 dose of study drug, and had a baseline SARS-CoV-2 viral RNA copy number ≥LLOQ.
LLOD, lower limit of detection; LLOQ, lower limit of quantitation; MMRM, mixed-effects model repeated measures; ODV, obeldesivir.

- At Day 5, a greater proportion of participants in the ODV group had negative SARS-CoV-2 nasal swab compared to the placebo group (nominal P = 0.001; Figure 3)

Figure 3. Proportion of Participants With Negative SARS-CoV-2 Nasal Swab Viral RNA Copy Number (Virology Analysis Set[a])

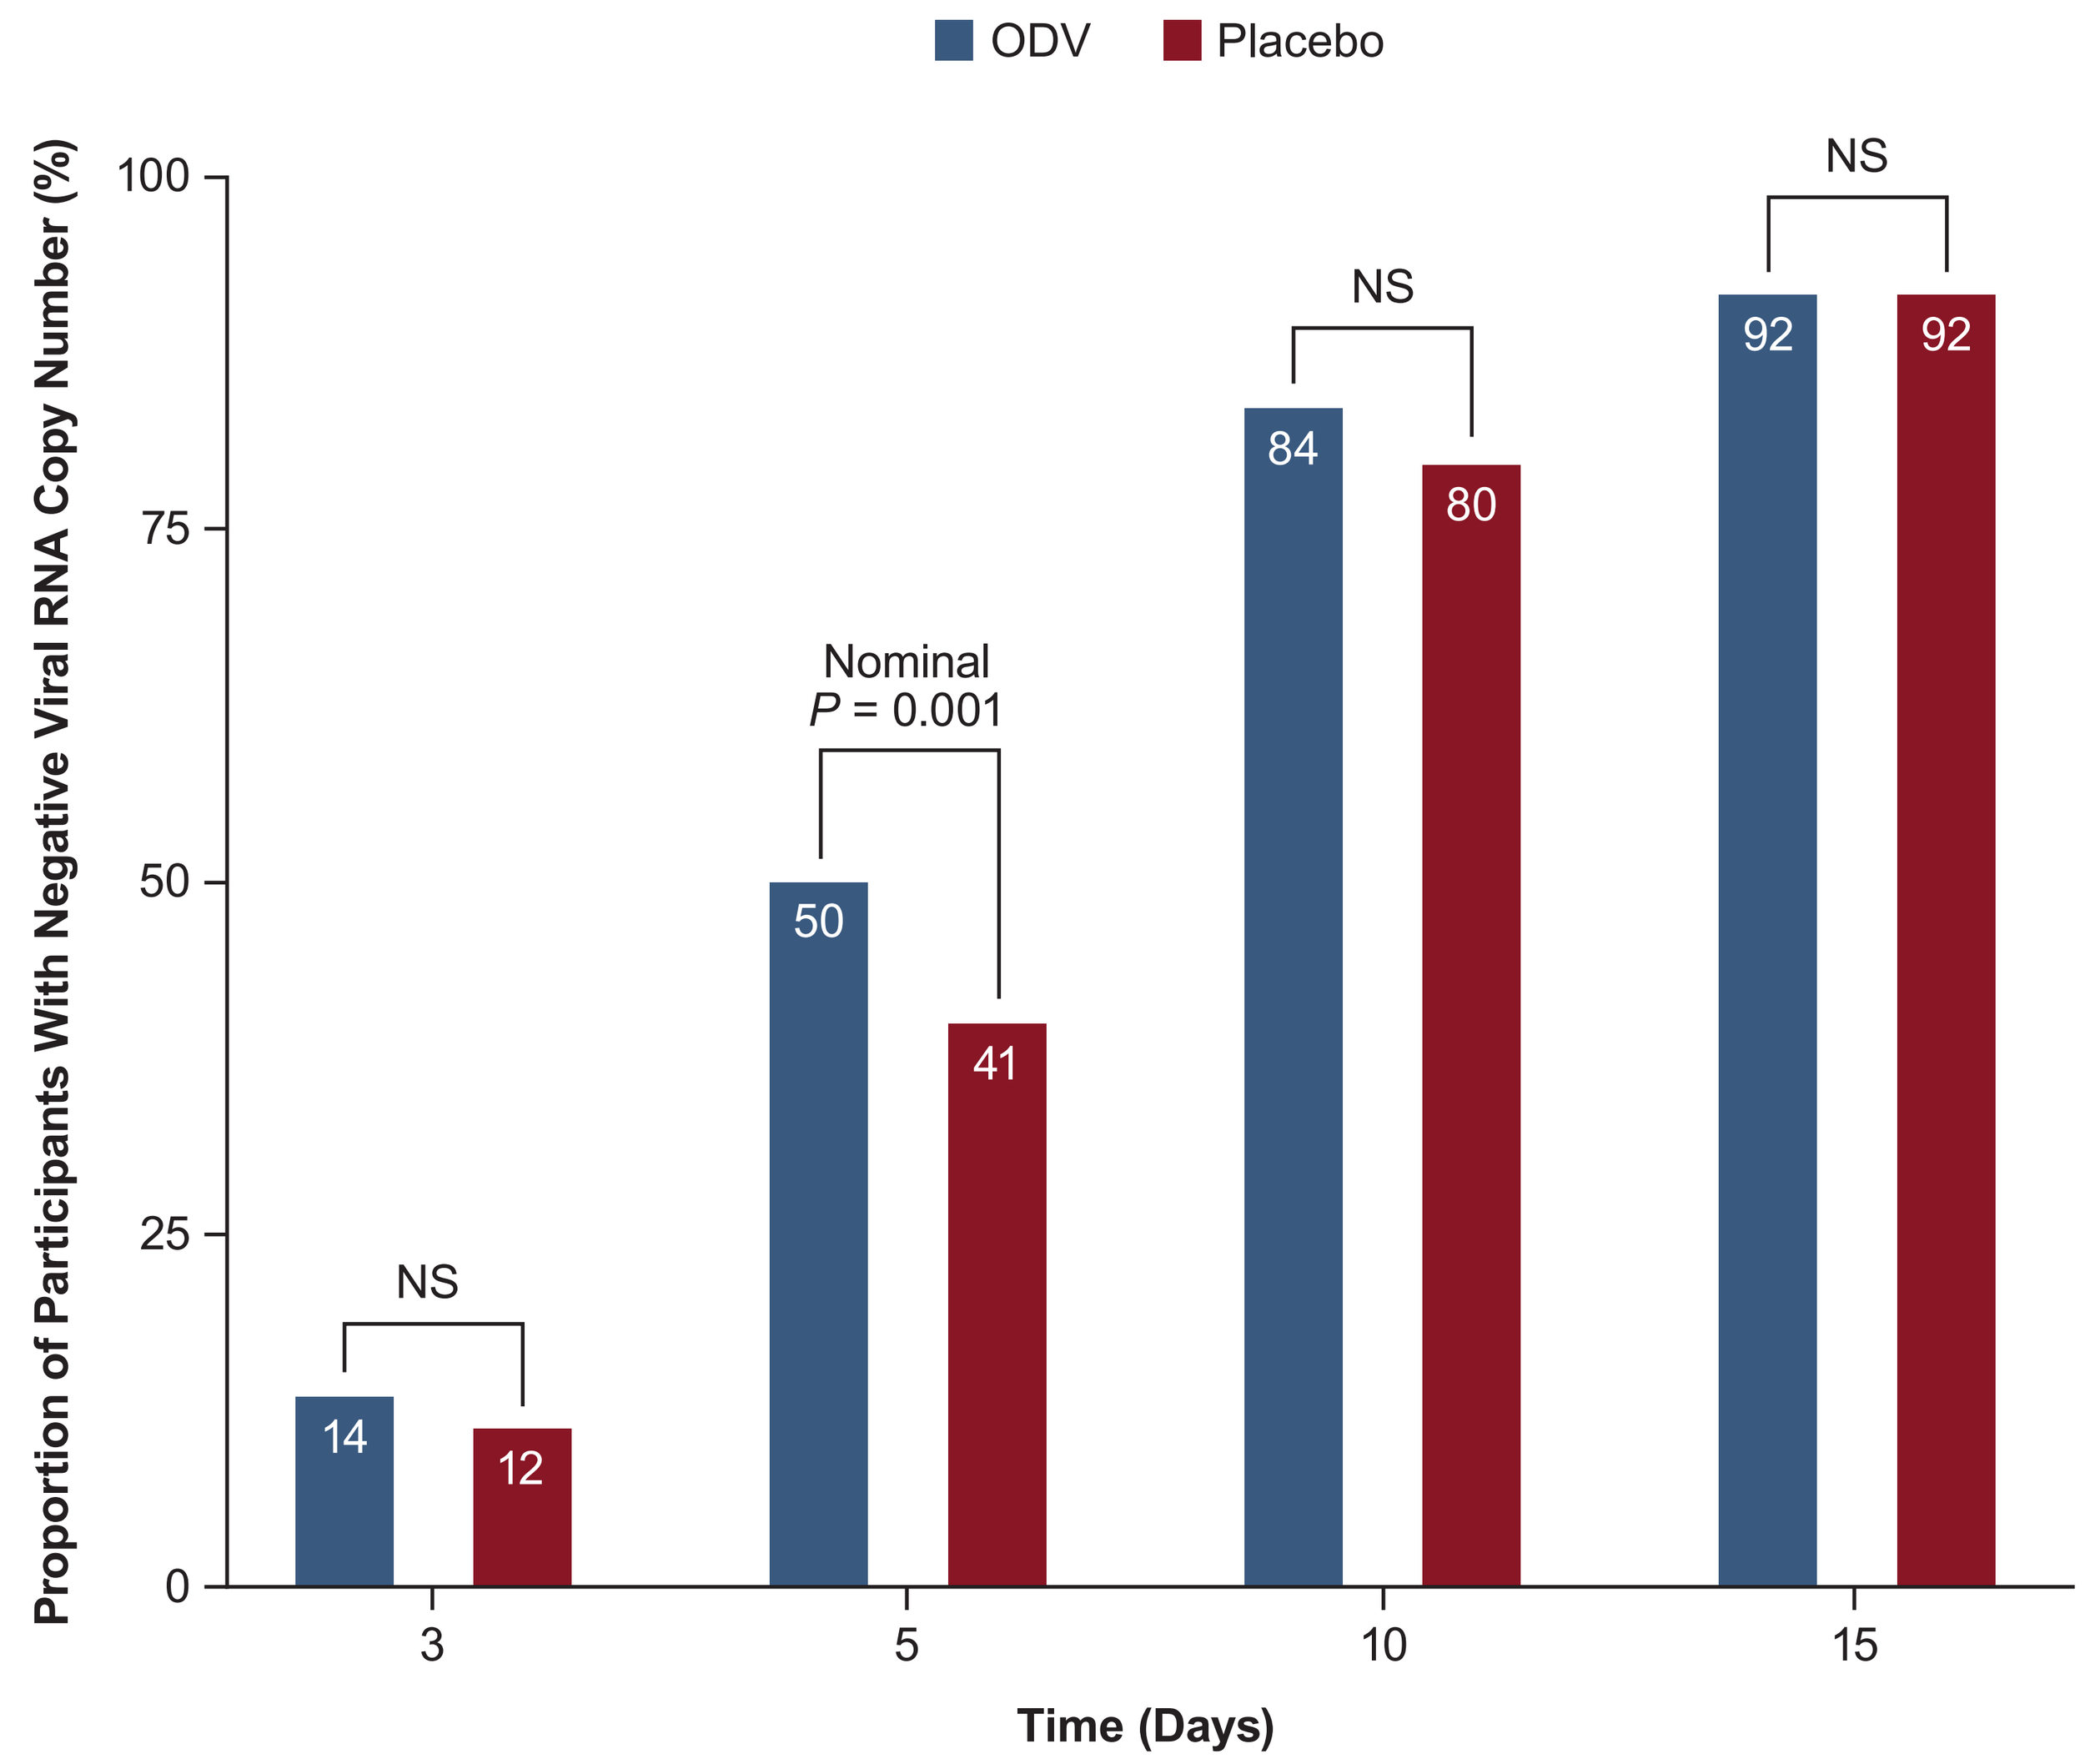

ODV
Placebo
Proportion of Participants With Negative Viral RNA Copy Number (%)
100
75
50
25
0
NS
14
12
Nominal
P = 0.001
50
41
NS
84
80
NS
92
92
3
5
10
15
Time (Days)

P values were from the Fisher's exact test.
[a]Virology analysis set included participants who were randomized, received ≥1 dose of study drug, and had a baseline SARS-CoV-2 viral RNA copy number ≥LLOQ.
LLOQ, lower limit of quantitation; NS, not significant; ODV, obeldesivir.

Safety

- The safety profiles of ODV and placebo were comparable, with similar rates of AEs, treatment-related AEs, serious AEs, and AEs leading to study drug discontinuation (Table 2)

Table 2. Incidence of Treatment-emergent AEs and Treatment-emergent Serious AEs (Safety Analysis Set[a])

n (%)	ODV (n = 979)	Placebo (n = 976)	Total (n = 1955)
Any AE	53 (5)	56 (6)	109 (6)
Grade ≥3 AE	2 (<1)	3 (<1)	5 (<1)
AE related to study drug	5 (1)	13 (1)	18 (1)
Grade ≥3 AE related to study drug	0	1 (<1)	1 (<1)
Serious AE	2 (<1)	4 (<1)	6 (<1)
Serious AE related to study drug	0	1 (<1)	1 (<1)
AE leading to premature discontinuation of study drug	1 (<1)	0	1 (<1)
Death	0	0	0

AEs were coded using the *Medical Dictionary for Regulatory Activities* Version 26.1. Severity grades were defined using the Division of AIDS Table for Grading the Severity of Adult and Pediatric Adverse Events Version 2.1 (July 2017).[11]
[a]Safety analysis set included participants who were randomized and received ≥1 dose of study drug. Participants in the safety analysis set were summarized according to the actual treatment received.
AE, adverse event; ODV, obeldesivir.

- There was a similar proportion of participants with ≥1 graded laboratory abnormality in the ODV (753/972 [77%]) and placebo (757/964 [79%]) groups; most (71%) laboratory abnormalities were Grade 1 or 2
 — Grade ≥3 laboratory abnormalities were reported in a similar proportion of participants across treatment groups (ODV, 59/972 [6%]; placebo, 82/964 [9%])

References: 1. Centers for Disease Control and Prevention. National Center for Immunization and Respiratory Diseases. The changing threat of COVID-19. Accessed June 26, 2024. https://www.cdc.gov/ncird/whats-new/changing-threat-covid-19.html. **2.** Centers for Disease Control and Prevention. COVID Data Tracker. Accessed August 14, 2024. https://covid.cdc.gov/covid-data-tracker. **3.** Centers for Disease Control and Prevention. Weekly U.S. influenza surveillance report. Accessed August 14, 2024. https://www.cdc.gov/flu/weekly/index.htm. **4.** World Health Organization. Therapeutics and COVID-19: Living Guideline. Accessed May 30, 2024. https://iris.who.int/bitstream/handle/10665/373975/WHO-2019-nCoV-therapeutics-2023.2-eng.pdf?sequence=1. **5.** Mackman RL, et al. *J Med Chem*. 2023;66:11701-17. **6.** Gottlieb RL, Paredes R. *Lancet Infect Dis*. 2024;24:108-10. **7.** Anoshchenko O, et al. *Clin Pharmacol Ther*. Published online June 28, 2024. doi:10.1002/cpt.3337. **8.** Amini E, et al. *Open Forum Infect Dis*. 2023;10:ofad500.574. **9.** Peng CC, et al. Presented at: 34th European Congress of Clinical Microbiology and Infectious Diseases (ECCMID); April 27-30, 2024; Barcelona, Spain. Poster 03881. **10.** Rodriguez L, et al. Presented at: IDWeek; October 11-15, 2023; Boston, MA, USA. Poster 545. **11.** Division of AIDS, National Institute of Allergy and Infectious Diseases, National Institutes of Health, US Department of Health and Human Services. Division of AIDS (DAIDS) Table for Grading the Severity of Adult and Pediatric Adverse Events. Corrected version 2.1. July 2017. Accessed August 19, 2024. https://rsc.niaid.nih.gov/sites/default/files/daidsgradingcorrectedv21.pdf.

Acknowledgments: We extend our thanks to the participants, their loved ones, and all participating investigators and site staff. This study was funded by Gilead Sciences, Inc. Medical writing and editorial support were provided by Catherine Bautista, PhD, of Lumanity Communications Inc., and were funded by Gilead Sciences, Inc.

Disclosures: **OO** served on advisory boards for Gilead Sciences, Inc., Moderna, and ViiV Healthcare/GSK. **JDG** consulted for Gilead Sciences, Inc., GSK, Invivyd, and Merck; received research support or grants from Gilead Sciences, Inc., Merck Sharp & Dohme (Biomedical Advanced Research and Development Authority), and Regeneron; and received nonfinancial support from Adaptive Biotechnologies, Labcorp, and Monogram Biosciences (outside of this study). **RLG** served as a consultant for AbbVie, AstraZeneca, Eli Lilly, Gilead Sciences, Inc., GSK, Johnson & Johnson, Roche Pharmaceuticals, and Roivant Sciences; served as a national coordinating primary investigator for Johnson & Johnson; served on an academic steering committee for Roivant Sciences; received from Gilead Sciences, Inc., a gift in kind to Baylor Scott & White Research Institute to facilitate NCT03383419; owns de minimis stock in AbCellera Biologics; and served as a speaker for Pfizer, outside the scope of COVID-19. **US** served on an advisory board for Gilead Sciences, Inc., and Regeneron; and received research support or grants from Pfizer. **MS** received research funding to conduct clinical trials from AstraZeneca K.K., Fujifilm Corporation, Genova Inc., Gilead Sciences, Inc., GSK K.K., Guerbet K.K., Insmed G.K., Kyorin Pharmaceutical Co., Ltd., Sanofi K.K., and Shionogi & Co., Ltd.; and received lecture fees from AstraZeneca K.K., GSK K.K., Kyorin Pharmaceutical Co., Ltd., and Sanofi K.K. **DNF** served on a Gilead Sciences, Inc., advisory board, outside the scope of COVID-19; served as a site primary investigator for clinical trials with Gilead Sciences, Inc., MetroBiotech, LLC, and Regeneron; and received investigator-initiated research support from Gilead Sciences, Inc. **EG** has business relationships with AstraZeneca, DBV, Genentech, Novartis, and Regeneron as a clinical research investigator. **PK** received institutional grants from Gilead Sciences, Inc., Merck, Theratechnologies, and ViiV Healthcare/GSK; received consulting fees from and served on an advisory board for Gilead Sciences, Inc., Merck, and ViiV Healthcare/GSK; and owns stocks/bonds in Gilead Sciences, Inc., Johnson & Johnson, Merck, Moderna, Pfizer, and ViiV Healthcare/GSK. **A Luetkemeyer** received research grant support to University of California San Francisco from Cepheid, Gilead Sciences, Inc., GSK, and Merck; and received laboratory support from Cepheid and Hologic. **A Lichtman**, **AM**, **YK**, **RHH**, **JL**, **AO**, **FD**, **LC**, **SD**, **CH**, **SC**, **KE**, and **PN** are employees of and may own stock or stock options in Gilead Sciences, Inc. **RH** is a former employee of and may own stock or stock options in Gilead Sciences, Inc. **AK** served on advisory boards for Gilead Sciences, Inc., and Madrigal Pharmaceuticals; served as a national coordinating principal investigator for Regeneron; and is a speaker for Madrigal Pharmaceuticals, outside the scope of COVID-19. **GA** has no conflicts to report.

IDWeek; October 16-19, 2024; Los Angeles, CA, USA